POSTER NUMBER: 24_TR

Multi-Center Analysis of Spontaneous Locomotor Activity in Pre-Clinical Models of Endometriosis using the HCA systems

Paulina Nunez-Badinez[1], Bianca De Leo[2], Nicole Schmidt[2], Philippa TK Saunders[3], Florent Barthas[4], Patrick Sweeney[5], Douglas J Armstrong[5,6], Rowland Sillito[5,6], Miguel Angel Tejada[7], Raul Gomez[7], Katy Vincent[8], Jens Nagel[1]

1. Bayer AG. Research & Development, Pharmaceuticals, Exploratory Pathobiology. Aprather Weg 18a, 42113 Wuppertal, Germany
2. Bayer AG. Research & Development, Pharmaceuticals, Reproductive Health. Müllerstr. 178, 13342 Berlin, Germany
3. Centre for Inflammation Research, The University of Edinburgh, Queen's Medical Research Institute, 47 Little France Crescent, Edinburgh, EH16 4TJ, United Kingdom
4. Grünenthal GmbH, 52099 Aachen, Germany.
5. Actual Analytics, 99 Giles St, Edinburgh, EH6 6BZ, United Kingdom
6. School of Informatics, University of Edinburgh, 10 Crichton St, Edinburgh, EH8 9AB United Kingdom
7. Instituto de Investigación Sanitaria INCLIVA, Valencia, Spain
8. Nuffield Department of Women's and Reproductive Health, University of Oxford, Oxford, United Kingdom

Introduction

Although a number of preclinical models for endometriosis (ENDO) have been described, readouts of spontaneous activity that could serve as surrogate measure of pain are understudied. A robust methodology to assess these readouts is important for translation of results in preclinical models into new and effective medicines for patients suffering from ENDO-associated pain (EAP).

In this study, we evaluated spontaneous activity in syngeneic transplantation models of ENDO using an in-cage monitoring system and compared results across four research institutions

Table 1. Refinements of the syngeneic model of ENDO. Please note that different histological features of the resulting lesions 3-4 weeks after model induction.

Endometriosis models in mouse: induction of peritoneal lesions and refinements

Donors - female mice: Balb/C ovaries intact cycling				
Basic model	Refinement 1 Syngeneic suture	Refinement 2 Syngeneic glue	Refinement 3 Syngeneic inoculation	Refinement 4 Syngeneic menses
Injection E2		Decidualised		Simulated menses
Uterine tissue - chopped or punch	Uterine tissue full thickness (punch)	Endometrial tissue (dec+basal), fragments	Endometrial tissue (dec+basal), homogenate	Endometrial tissue dec+basal+shed homogenate
Recipients - female mice: ovaries intact cycling. Insertion of microchips compatible with HCA equipment				
Tissue pieces introduced into peritoneal cavity Lesions (cysts) 3 weeks	Tissue pieces stitched onto peritoneal wall & mesentery Lesions (cysts) 3-4 weeks	Tissue fragments glued onto peritoneal wall + E2 priming on recipients Lesions (mix histology) 3-4 weeks	Tissue homogenate injected into peritoneal cavity Lesions (mix histology) 3-4 weeks	Endometrial homogenate i.p injection Lesions 3-4 weeks, no cysts, immune cells, nerves
Data gathering in home cage analysis system				
Data analysis packages and unbiased evaluation				

Methods

Experiments were done in a multi-centre approach following strict compliance to local authorities on animal experimentation. The syngeneic tissue in-transplantation model was implemented in BALB/c mice at 8-12 weeks old. Refinements of the basic model were perfromed as described (Table 1), and the impact of these refinements were compared to evaluate how these affected the spontaneous activity of the animals, which were recorded using the Home Cage Analysis (HCA) system. Data was processed for each individual (identified by ID of implanted chip); then grouped by treatment group (ENDO or SHAM), and finally grouped by refinement used over time. Activity over time was corrected for overall baseline activity.

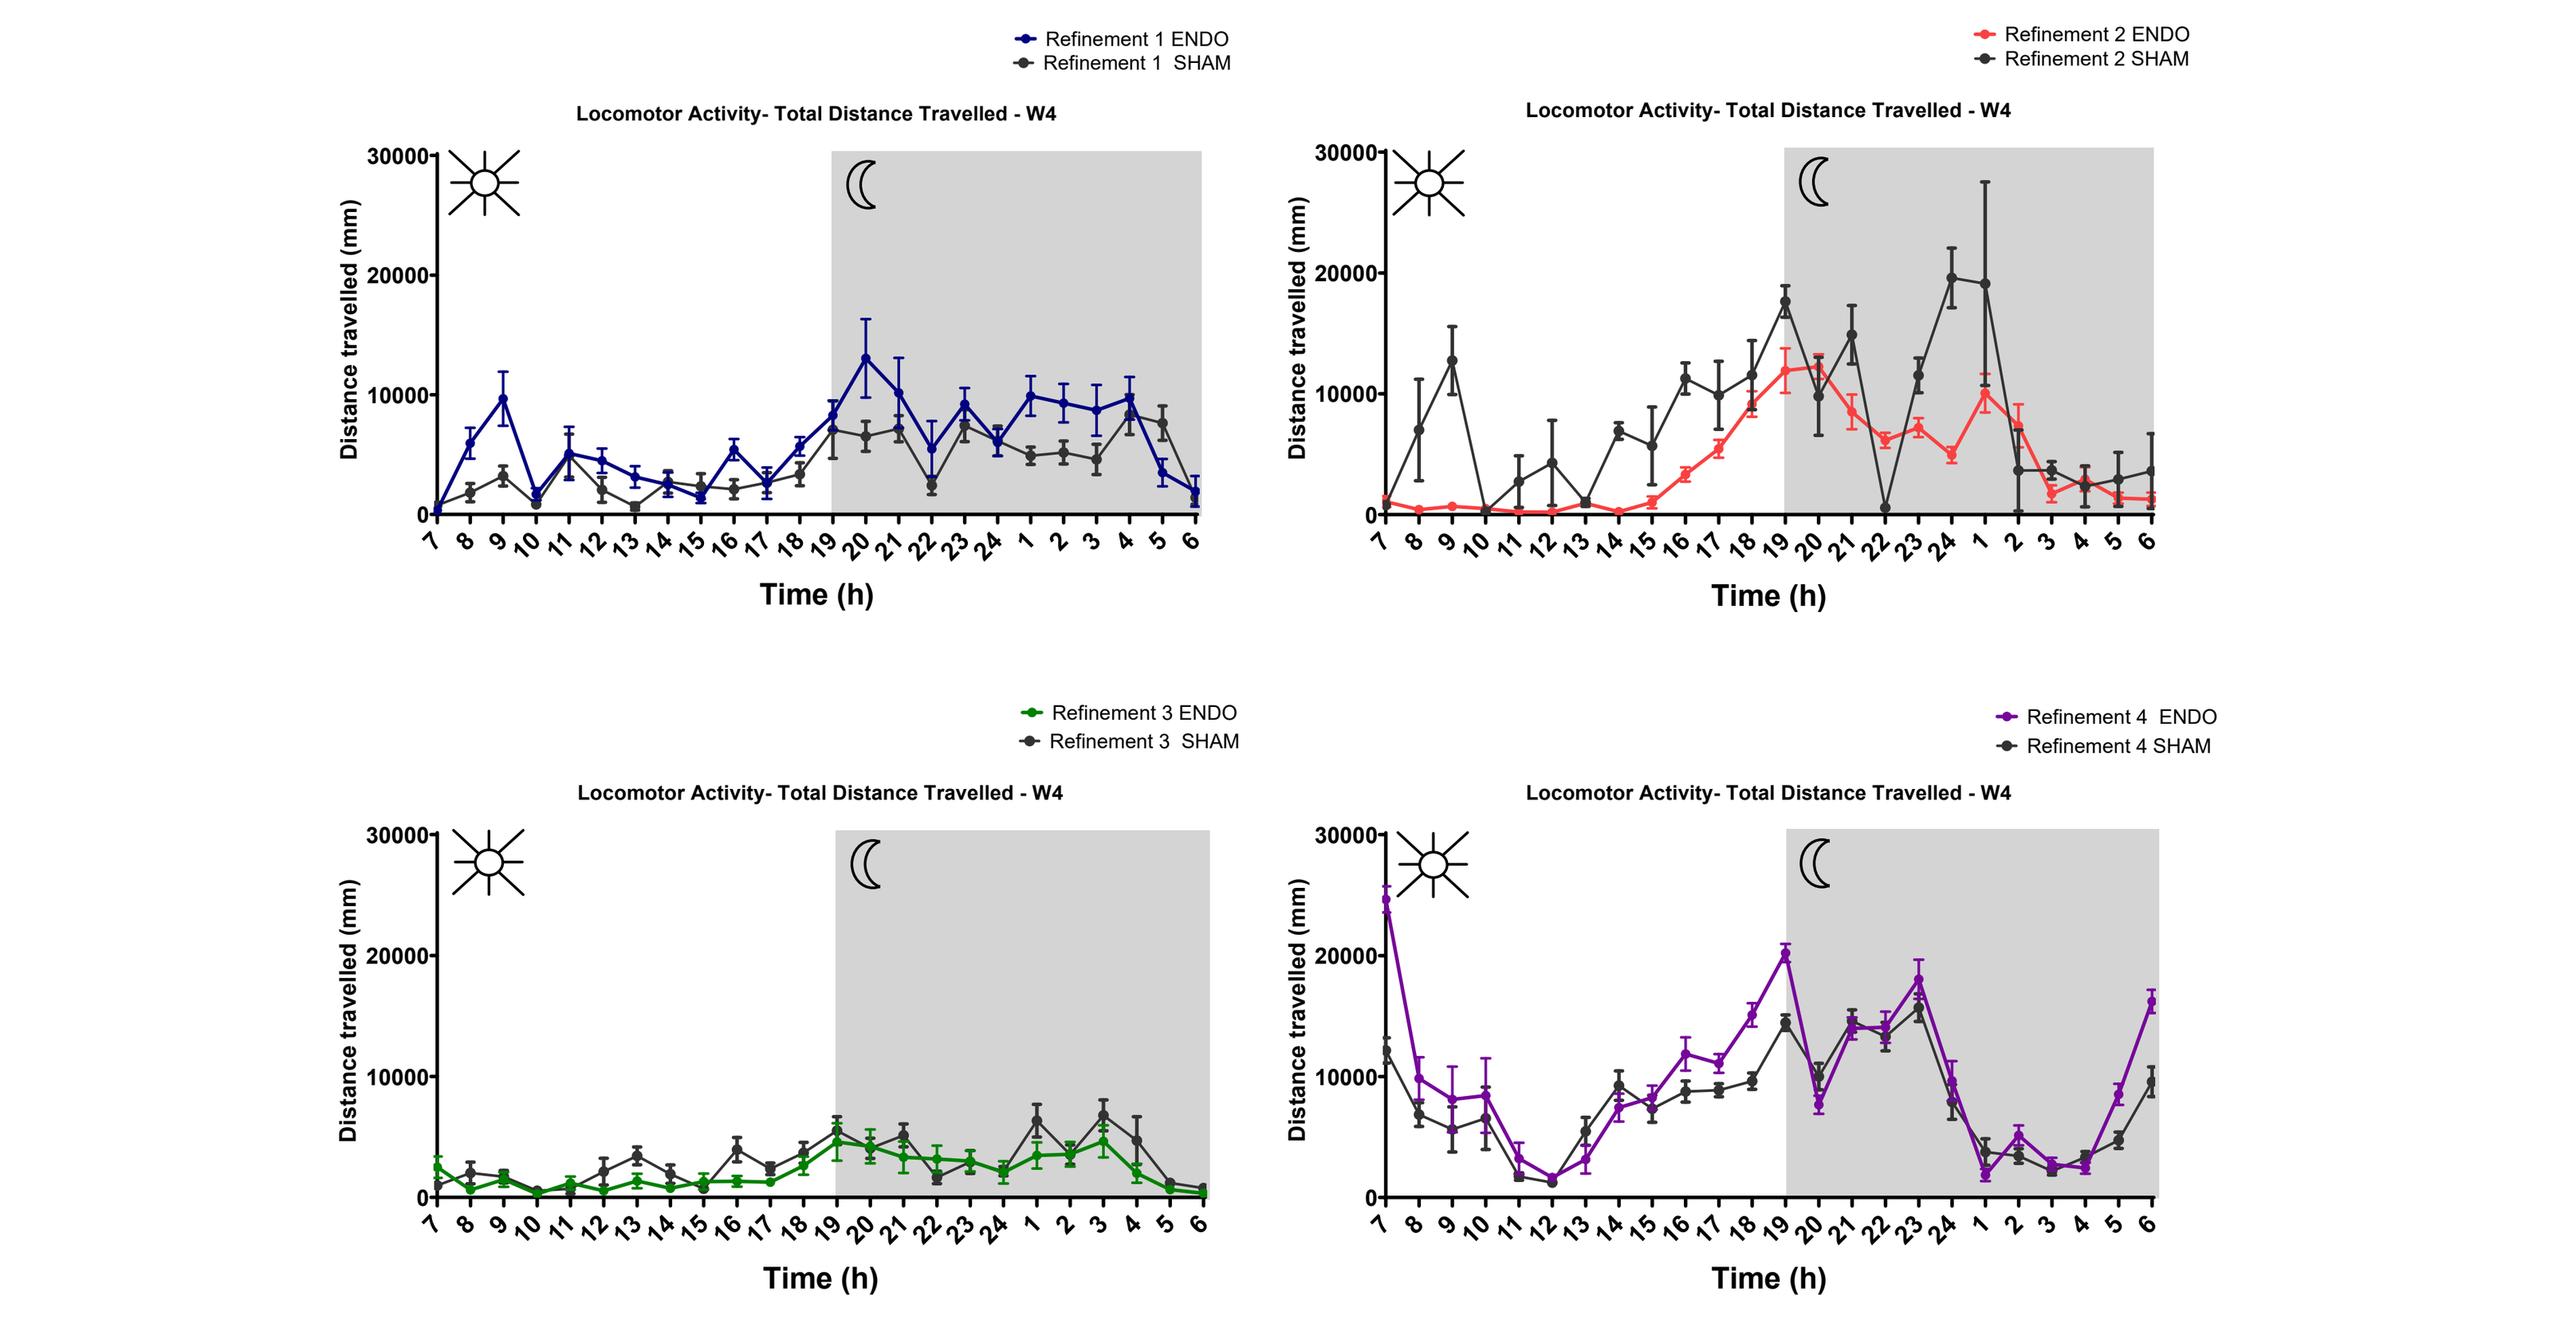
Refinement 1 ENDO
Refinement 1 SHAM
Refinement 2 ENDO
Refinement 2 SHAM
Refinement 3 ENDO
Refinement 3 SHAM
Refinement 4 ENDO
Refinement 4 SHAM
Locomotor Activity- Total Distance Travelled - W4
Distance travelled (mm)
Time (h)

Figure 1. Locomotor activity in in a 24-h timeframe of ENDO model refinements. Example plots of the activity levels at week 4 after model induction. Please note the spikes in activity levels during the first hours after lights go on in refinements 1, 2 and 4, as well as when lights go off. These effects were observed in all timepoints measured. Further analyses of activity considered the following time windows: 1) 10 am – 5 pm as daytime, 2) 10 pm- 5 am as night-time, and 3) 5-6 am as anticipation daytime.

1. Identification of relevant time windows for analysis of spontaneous activity

In an initial analysis, the spontaneous locomotor activity per hour in a 24h timeframe was calculated (Figure 1). External environmental stimuli impacted the spontaneous activity patterns of mice. For example, during the first hours after lights go on (7-10 am), most mice responded to the presence of staff undertaking standard tasks. Also, the last hour before lights going on appeared to be a sensitive time. We refined our evaluation to compare following time windows of relevance: 10 am - 5 pm (light phase), 10 pm – 5 am (dark phase), and we included separately the last hour of the dark phase: 5-6 am, and considered as anticipation for lights going on.

Results

2. Spontaneous locomotor activity of syngeneic model variants of ENDO

Generally, activity levels of animals increased during night-time independent of intervention or research centre. In refinement 1, mice from the ENDO group displayed higher activity levels than SHAM during weeks 1-3 after model induction. This effect was observed during all time windows tested (Figure 2 A, E & I). Within the ENDO group the activity levels were strongly reduced during week 1 (W1, about 50% of baseline activity), and appeared to stabilize starting from W2 onwards. In refinement 2, the ENDO group displayed a sustained decrease in spontaneous activity compared to SHAM at all time windows (Fig 2B, F & J). These differences were less pronounced during anticipation time and closer to W3, although being again pronounced at W4. In refinement 3, the ENDO group showed decreased activity levels compared to SHAM at W4 during daytime (Fig 2C), but not in the other time windows (Fig 2G & K). In refinement 4, no differences were found in activity levels between ENDO and SHAM groups during daytime and nighttime (Fig 2D & H), but strong differences between groups at anticipation for daytime (Fig 2L) were observed. It is unclear if these differences are due to a biological effect or to a so-called in-cage effect, since n=4 mice/cage/group were tested.

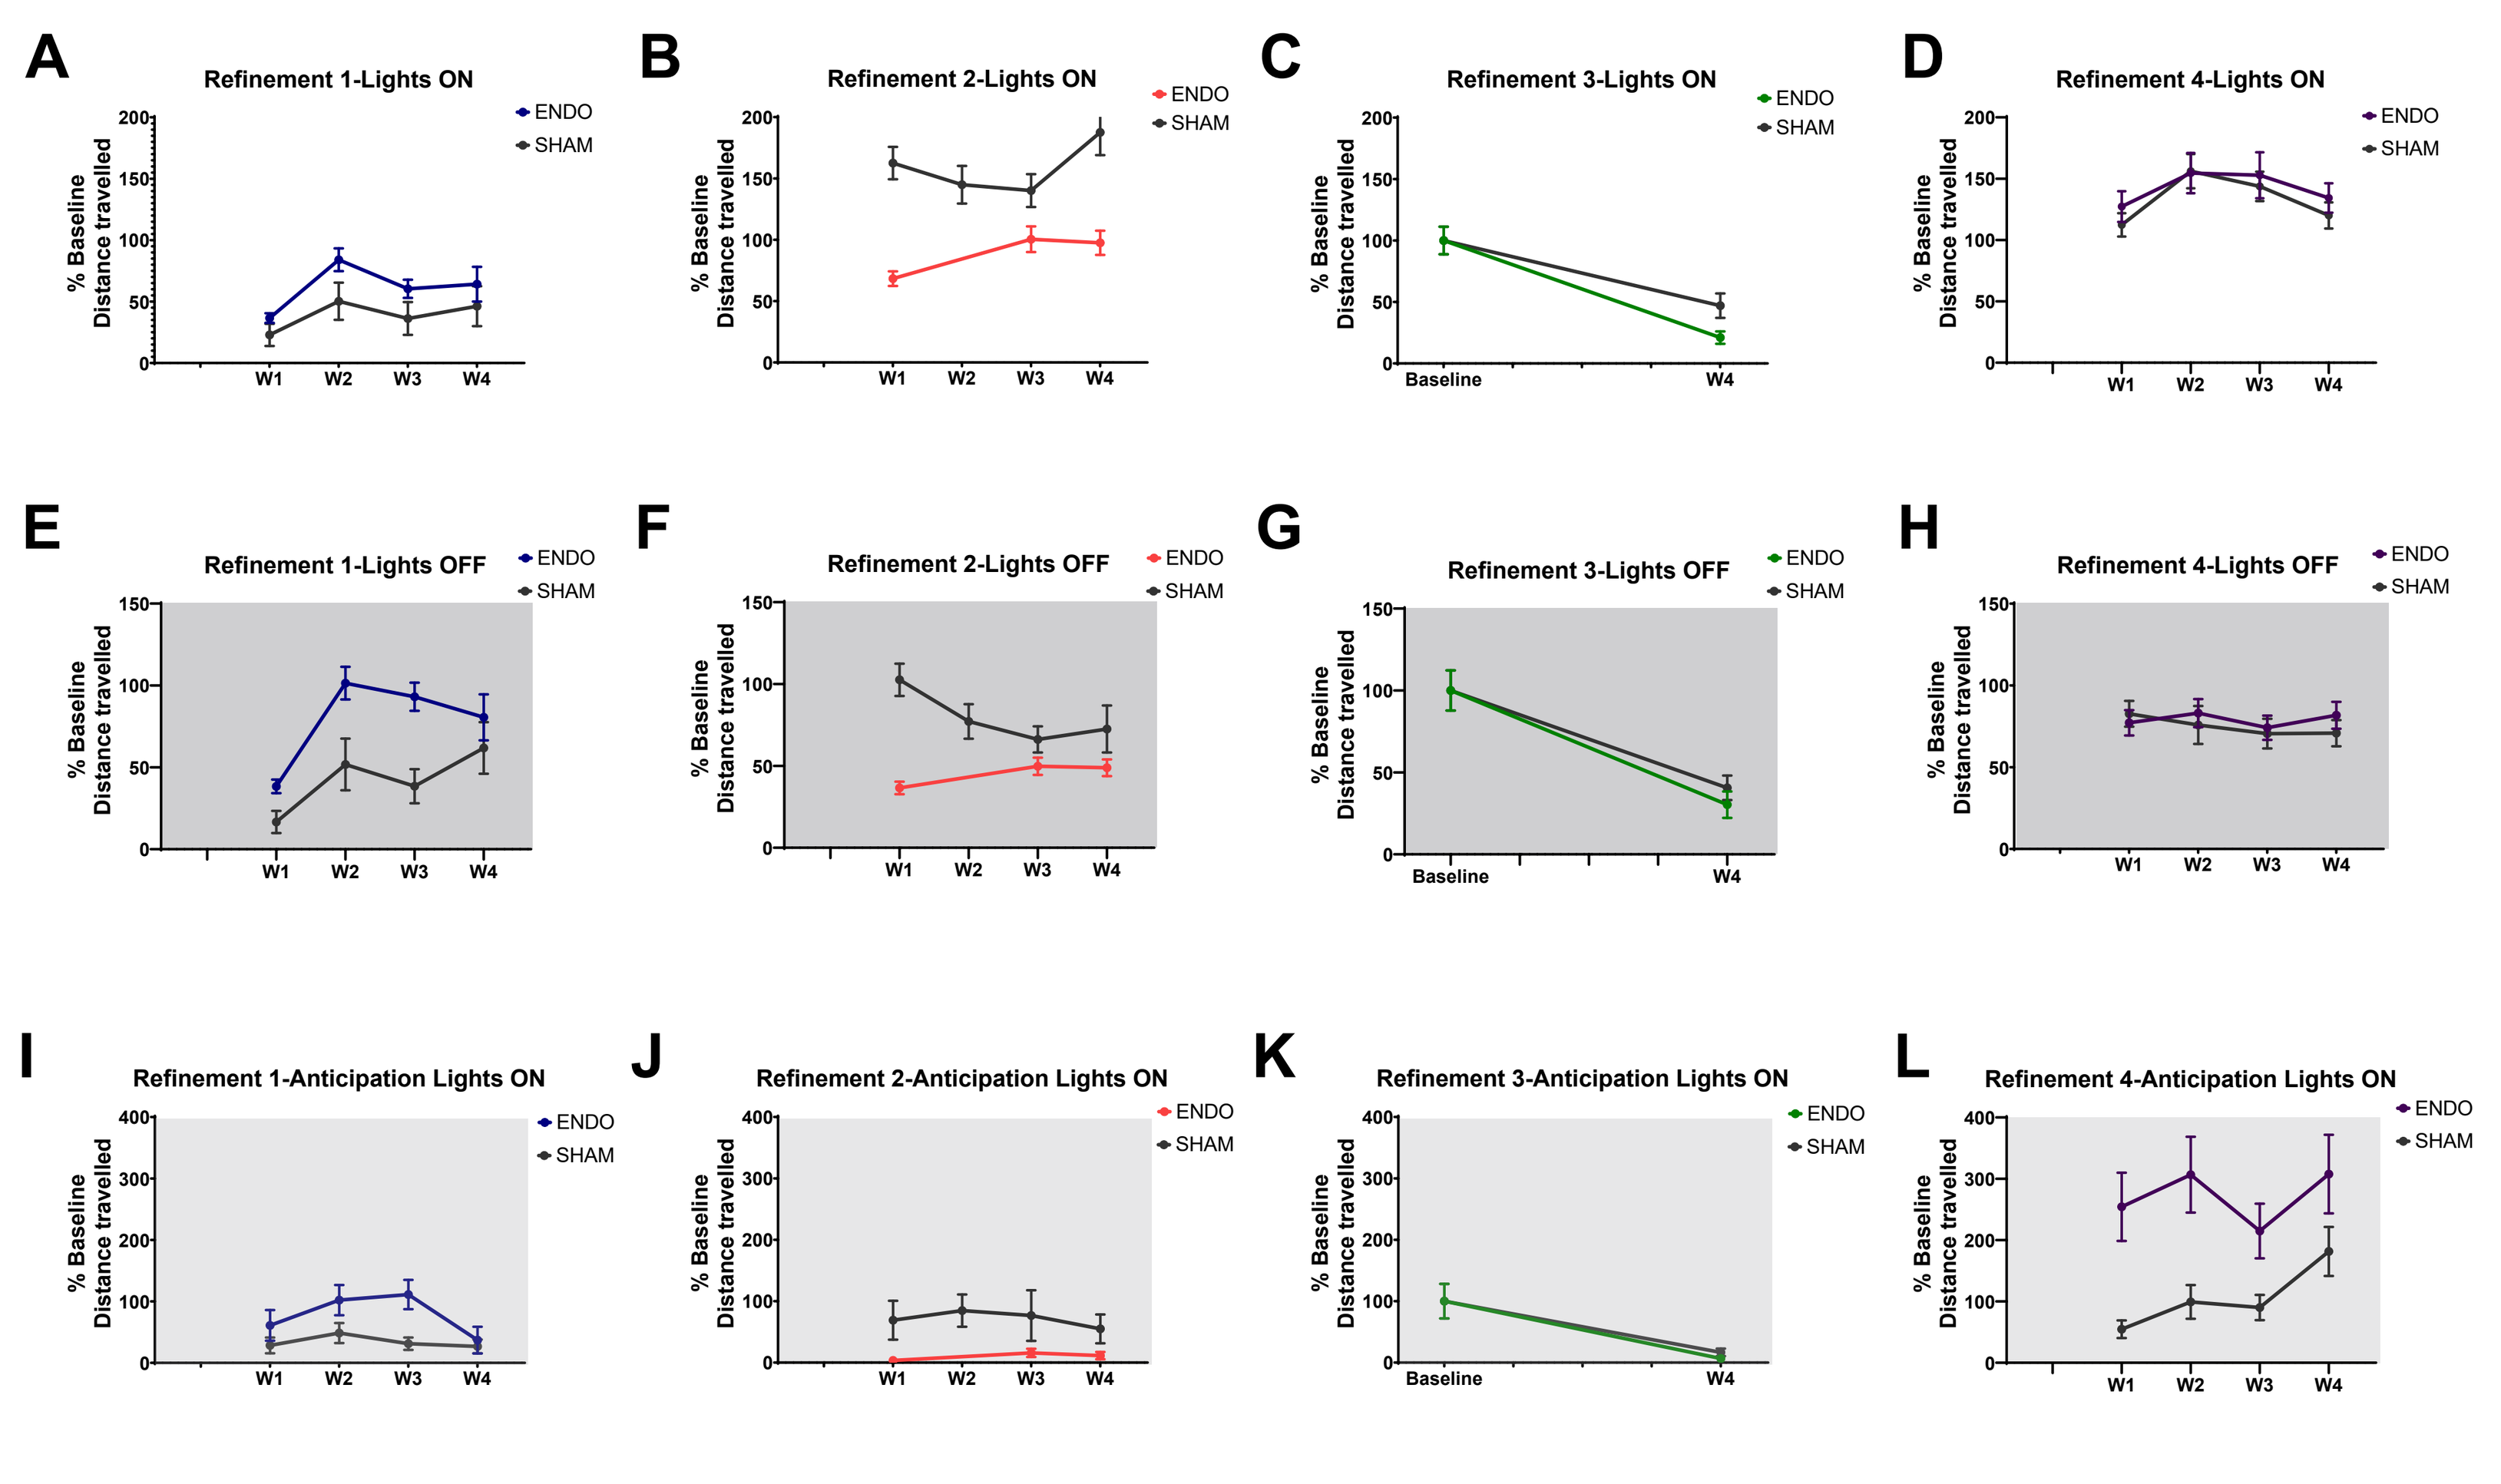
A
Refinement 1-Lights ON
B
Refinement 2-Lights ON
C
Refinement 3-Lights ON
D
Refinement 4-Lights ON
E
Refinement 1-Lights OFF
F
Refinement 2-Lights OFF
G
Refinement 3-Lights OFF
H
Refinement 4-Lights OFF
I
Refinement 1-Anticipation Lights ON
J
Refinement 2-Anticipation Lights ON
K
Refinement 3-Anticipation Lights ON
L
Refinement 4-Anticipation Lights ON
ENDO
SHAM
% Baseline Distance travelled
W1
W2
W3
W4
Baseline

Figure 2: Spontaneous locomotor activity during three relevant time windows for analysis. Summary of locomotor activity (presented as percentage of baseline activity, mean ± SEM). A-D: during Lights ON (10 am-5 pm), E-H: during Lights OFF (10 pm-5 am) & I-L: during Anticipation lights ON (5-6 am). A, E & I : Refinement 1. B, F & J: Refinement 2. C, G & K: Refinement 3. D, H & L: Refinement 4

3. Outlook

This pilot study and analysis provided us first insights on the impact of our model refinements on spontaneous beahviours using the HCA systems. Locomotor activity is only one readout of several other behaviors recorded, and these will be incorporated into the final manuscript. Additionally, we are performing H&E and cytokeratin stainings of the lesions developed in these animals in order to compare their histological characteristics (data in preparation). We are planing a new multi-center experimental run that will reproduce the syngeneic glue model in three research centers with an increased n and longer recording times to increase statistical power.

Conclusions

The use of in-cage monitoring to record spontaneous behaviours avoids operator bias and is considered more relevant to patient experience. In particular, spontaneous locomotor activity reflects the ongoing distress of the animals and appeared to be differentially affected in syngeneic model variants for ENDO. Some methodologies affected both ENDO & SHAM groups.

This study shows the importance of cross-site comparison of methods which has revealed variations in behavior in validated preclinical models of ENDO including environment. We strongly reccommend to record these variations as part of experimental protocols.

In future experiments we will refine the use of this tool so that we can characterise this and other pain components in the syngeneic glue model refinement of ENDO based on a back-translational approach.

Acknowledgements

This project has received funding from the Innovative Medicines Initiative 2 Joint Undertaking under grant agreement No 777500. This Joint Undertaking receives support from the European Union's Horizon 2020 research and innovation programme and EFPIA Companies.

The statements and opinions presented here reflect the author's view and neither IMI nor the European Union, EFPIA, or any Associated Partners are responsible for any use that may be made of the information contained therein.

© Bayer | Research & Development, Pharmaceuticals | Paulina Nunez-Badinez | 2022

www.imi.europa.eu; www.imi-paincare.eu